CANCER SUPPORT COMMUNITY
COMMUNITY IS STRONGER THAN CANCER

ACADEMY OF ONCOLOGY NURSE & PATIENT NAVIGATORS (AONN+) CONFERENCE, NOVEMBER 2021

For more information: 1-888-793-9355; **www.CancerSupportCommunity.org**
Contact: Claire Saxton, MBA: csaxton@CancerSupportCommunity.org

Patient Preferences and Understanding of a Precision Medicine Lexicon—Towards the Development of Patient-Friendly, Consistent Terminology

M. Claire Saxton, MBA[1], Maria B. Gonzalo, MS[1], Jenny Karubian, MA[2], Helen M. Nichols, PhD[1], Sanjana Quasem, MPH[1], Danelle Johnston, MSN, RN, HON ONN CG, OCN[1]
[1]Cancer Support Community, Washington, DC, [2]Ready to Launch Research, Los Angeles, CA

BACKGROUND

As precision medicine testing and treatment options evolve, the complexity of what cancer patients and caregivers need to understand to be active members in their health care team has increased dramatically. Patients and caregivers are overwhelmed and confused by precision medicine terms and explanations of concepts that can vary significantly from provider to provider, within and across institutions.

Developing clear, consistent precision medicine terminology is critically important to ensure patients are making informed decisions and receiving optimal care. With this goal in mind, CSC developed a precision medicine lexicon that health care providers, patients, oncology professional associations, industry, and patient advocacy organizations can mutually adopt.

OBJECTIVE

Building on the work of the Consistent Testing Terminology Working Group (**www.CommonCancerTestingTerms.org**), CSC aimed to co-create with providers, patients, and caregivers a lexicon related to precision medicine, biomarker testing, and genetic testing to improve patient and caregiver understanding and patient/provider communication.

METHODS

CSC's precision medicine lexicon was developed with input from oncology professionals and then subject to iterative focus group and online discussion board testing with patients and caregivers. The research captured feedback and insights regarding clarity, familiarity, preference for terms used, and overall understanding of precision medicine terms to use in the lexicon. Clarity, understanding, and preference for terms was validated using an online survey. Respondents were selected to be representative of US cancer patients and caregivers in gender, race/ethnicity, and household income. Blacks and Hispanics were oversampled to test differences by race and ethnicity. The general public was sampled to validate the lexicon with people who were diagnosis-naive.

CONCLUSIONS AND IMPLICATIONS

CSC's Precision Medicine Lexicon incorporates patient and caregiver preferences & insights and is validated as easy to understand by patients, caregivers, & members of the general public. While work remains to be done to help even more people with low health literacy understand precision medicine & biomarker testing concepts, adoption of this lexicon can help bridge communication gaps between most patients and their health care teams. The final lexicon is available for health care providers, patient advocacy groups, and industry to use precision medicine terms that are patient- and caregiver-preferred and definitions that patients and caregivers have indicated are easy to understand. **www.CancerSupportCommunity.org/PMPlainLanguage**

SURVEY RESPONDENT CHARACTERISTICS

Total sample size: 614
~ 50% Cancer patients and/or caregivers
~ 50% General population

Average Age: 46.7 years old (SD=18.1 years)

Gender: ~ 50% Female

Race/ethnicity: 69% White / 21% Black / 6% Asian or South Asian / 0.3% American Indian / 2% multiracial / 2% Other / 14% Hispanic

Household Income: 36% <$50K / 34% $50K-$100K / 30% >$100K

Education: 3% <HS / 44% HS or some college / 53% Bachelor's+

SURVEY RESULTS

CONCEPT AS DEFINED IN LEXICON	CLARITY SCORE*
Genes	90%
Inherited Mutation	87%
Genetic Testing for Inherited Cancer Risk	86%
Genetic Testing for an Inherited Mutation	83%
Biomarker	83%
Acquired Mutation	81%
Biomarker Testing	84%
Cancer Subtypes	81%
Liquid Biopsy	81%
Tissue Biopsy	85%

**Clarity Score = % of respondents who gave a score of 7 or greater out of 10*

Lexicon definitions were scored by most survey respondents as highly understandable (clarity score of 7 or greater out of 10). There were no significant differences in clarity scores between patients/caregivers and the general public. Reporting Hispanic ethnicity was associated with significantly lower (up to 14%) clarity scores. Lower household income (<$50k) and lower education attainment (<Bachelor's degree) were associated with significantly lower (up to 12%) clarity scores for all but 2 terms: Genes & Tissue Biopsy.

INHERITED MUTATION/ GENE MUTATION

The top terms for inherited mutations that respondents said they understood and somewhat or strongly favored were **inherited mutation, gene mutation, and genetic mutation**. However, genetic mutation was significantly less favored by Blacks than whites. Notably, pathogenic variant was the least understood and the least preferred term of 13 offered.

GENETIC TESTING FOR INHERITED CANCER RISK

Respondents favored the term **genetic testing for inherited cancer risk** when referring both to testing people who have not been diagnosed with cancer but have a family history (69%) and referring to testing people already diagnosed with cancer (69%).

GENE MUTATION/ GENETIC MUTATION/ BIOMARKER

The top terms for acquired mutations that respondents said they understood and favored were **genetic mutation** (this term was favored significantly more by Blacks than whites) and **gene mutation** (favored significantly less by Blacks than whites). The top terms favored by respondents that make a clear distinction between inherited and acquired mutations were **cancer marker** and **biomarker** (but both were favored significantly less by Blacks than whites).

Blacks tended to report higher familiarity and understanding of terms than whites. Caregivers tended to report higher familiarity and understanding of terms than patients. Patients & caregivers together tended to report higher familiarity and understanding of terms than the general public. Respondents with lower household income and education attainment were significantly less likely to report familiarity, understanding, and preference of terms.

BIOMARKER TESTING/ GENE-BASED CANCER TESTING/ GENETIC TESTING OF THE CANCER/ COMPREHENSIVE BIOMARKER TESTING

While there is a multistakeholder effort to use the term **biomarker testing** as an umbrella term (favored by 55% of respondents), more specific terms are needed to distinguish genomic testing from other biomarker testing. The most favored terms were:

- **Gene-based cancer testing** (59%, no difference by income)
- **Genetic testing of the cancer** (58%, no difference by income or education)
- **Cancer marker testing** (55%, no difference by income)
- **Comprehensive biomarker testing** (54%, less preferred by Hispanics).

FRANKLY SPEAKING ABOUT CANCER
A PROGRAM OF THE CANCER SUPPORT COMMUNITY

Frankly Speaking About Cancer: Precision Medicine Program

CSC's FREE educational program, *Frankly Speaking About Cancer: Precision Medicine*, utilized this research to provide easy-to-understand information for patients & caregivers as they navigate precision medicine, biomarker testing, and treatment decisions. **www.CancerSupportCommunity.org/Precision-Medicine**.

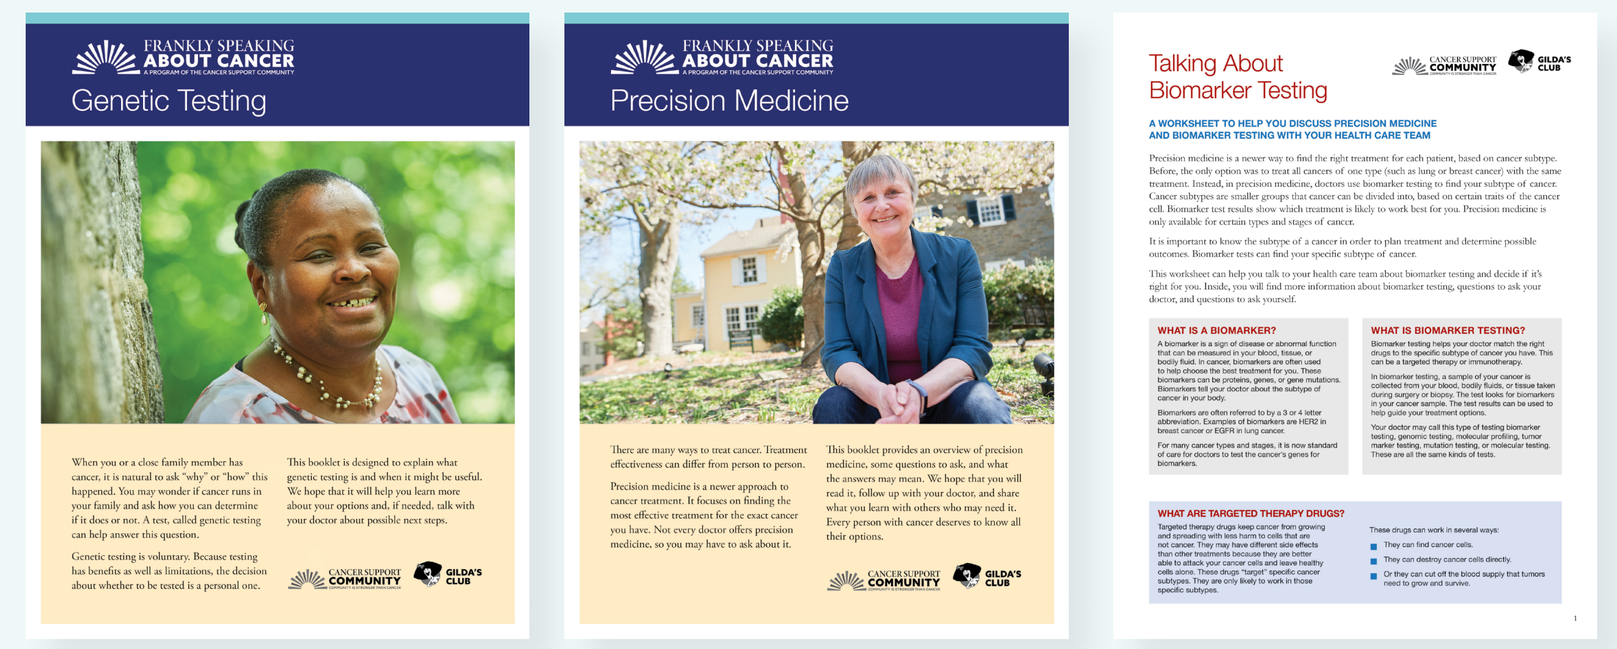
Genetic Testing
Precision Medicine
Talking About Biomarker Testing

ACKNOWLEDGMENTS: This project was supported by grants from AbbVie, Amgen Oncology, Bristol Myers Squibb, Exact Sciences, Foundation Medicine, Genentech, Lilly Oncology, Merck, NeoGenomics, Takeda.

REFERENCES: Ciardiello, F., Adams, R., Tabernero, J., Seufferlein, T., Taieb, J., Moiseyenko, V., Ma, B., Lopez, G., Vansteenkiste, J. F., Esser, R., & Tejpar, S. (2016). Awareness, Understanding, and Adoption of Precision Medicine to Deliver Personalized Treatment for Patients With Cancer: A Multinational Survey Comparison of Physicians and Patients. *The Oncologist*, 21(3), 292–300. https://doi.org/10.1634/theoncologist.2015-0279; Roundtable on Health Literacy, Board on Population Health & Public Health Practice, Health & Medicine Division, National Academies of Sciences, Engineering, & Medicine. (2016). Relevance of Health Literacy to Precision Medicine: Proceedings of a Workshop. National Academies Press (US). Wynn, R. M., Adams, K. T., Kowalski, R. L., Shivega, W. G., Ratwani, R. M., & Miller, K. E. (2018). The Patient in Precision Medicine: A Systematic Review Examining Evaluations of Patient-Facing Materials. *Journal of Healthcare Engineering*, 2018, 9541621. https://doi.org/10.1155/2018/9541621